ADVERSE REACTION TO PARENTERAL AMINO ACIDS (AA) IN A PATIENT WITH MAST CELL ACTIVATION SYNDROME (MCAS) – A CASE REPORT

Angelina Mason, RD, LDN, CNSC, BriovaRx Infusion Services, Columbia, Maryland, UNITED STATES

Background:

Mast cells are a part of IgE mediated inflammation. They are found throughout the body, in particular in mucosal membranes, lungs, skin, and the GI tract. Mast cells contain histamine, and are effectors in anaphylactic reactions. MCAS is a condition where mast cells can have an abnormal release of inflammatory mediators, leading to a constellation of systemic reactions, including anaphylaxis. Treatment frequently involves the administration of antihistamines and avoiding triggers. Dietary recommendations include avoiding foods with high levels of histadine (His), the AA precursor of histamine.

Methods:

KW is a 17 year old female diagnosed with MCAS in 2017. KW was referred to a national home infusion company for initiation of home parenteral nutrition (PN) in 2016 related to weight loss, poor oral intake, delayed intestinal transit time and poor motility. She presented at 51.4kg, 165cm, which was 73% of her usual body weight (UBW). KW was started on a lipid free PN formula of 200 gm of dextrose, 50 gm of AA (Travasol®), electrolytes, MTE concentrate and MVI in 1500 ml, starting on a 24 hour infusion with plans to cycle and advance macronutrients to meet nutritional needs. After three weeks of PN, KW experienced urinary retention, abdominal pain, and edema with PN infusions. MVI was held, but symptoms persisted. PN was discontinued after 5 weeks of therapy since symptoms continued to worsen. She was then maintained on dextrose-based hydration.

KW restarted HPN in 2017 due to weight loss after a vascular procedure, with weight down to 48.6 kg. The physician requested a "high protein" formula. Lipid free PN was initiated at 1000 ml, 200 gm of dextrose, 80 gm of AA (Prosol®) over 24 hours with electrolytes, MVI and MTE concentrate. Eight hours into the infusion, patient started complaining of headache, flushing, tachycardia, shortness of breath, and feeling tightness in her throat. PN infusion was stopped, and antihistamines administered. Symptoms subsided for several hours. Antihistamines were administered again after about 6 hours. PN was held, and symptoms resolved. Patient has been maintained on dextrose containing hydration with MVI since.

Discussion:

All parenteral AA solutions contain His, a non-essential AA in varying concentrations. (Table 1.) Histadine converts to histamine in a one-step decarboxylation reaction. (Figure 1.) KW had received parenteral infusions with dextrose and MVI without any adverse reactions. It was only with the addition of AA that symptoms of intolerance arose. When KW received PN in 2016, she did not yet have a diagnosis of MCAS, but her symptoms were consistent with the syndrome. Given this diagnosis, it is possible that adverse reactions to AA could be related to the dose of His in the PN, especially considering the milder reaction occurred with a dose of 2400 mg, and a severe reaction in 2017 occurred with a 4720 gm dose.

Conclusion:

PN is a complex therapy that may contain up to 30-40 different ingredients. Reactions to individual components in PN are rare and typically are associated with IV lipid emulsions, and less frequently parenteral MVI. Reactions to parenteral AA are infrequent, but can occur in patients with inborn errors of metabolism, or other conditions that affect amino acid metabolism. Careful initiation and monitoring for adverse reactions to PN is an essential responsibility of the nutrition support team.

Table 1.

Solution	Concentration (%)	gm AA per 100ml	mg His per 100ml	mg His per 1gm AA
Travasol	10	10	480	48
FreAmine III	10	10	280	28
Clinisol	15	15	894	59.6
Prosol	20	20	1180	59
Aminosyn II	10	10	300	30
Aminosyn II	15	15	450	30

Figure 1.

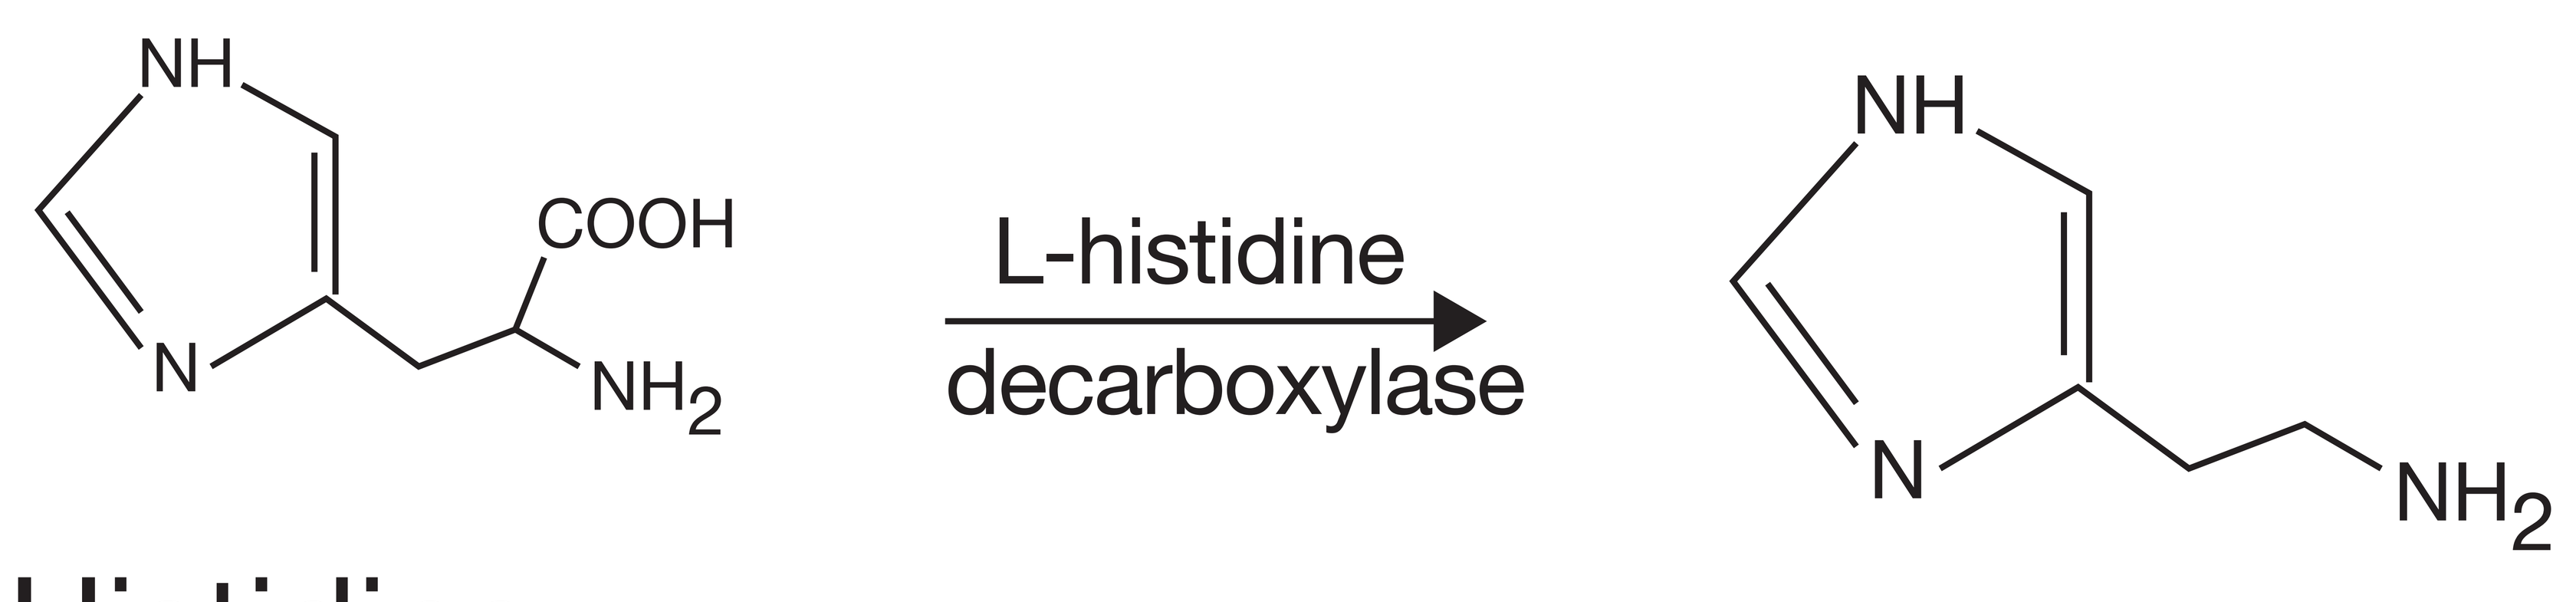
NH
N
COOH
NH_2
L-histidine
decarboxylase
NH
N
NH_2

Histidine

Histamine